Worldwide Clinical Trials

Obesity Assessments
Clinical Meaningfulness vs. Operational Complexity

Peyton Sandroni, PhD
Assistant Director, Clinical Research Methodology

MarieElena Cordisco, MA, NP-C, APRN
Senior Director Therapeutic Strategy Lead, Cardiometabolic

Natalia Castro
Executive Director, Project Management Cardiometabolic

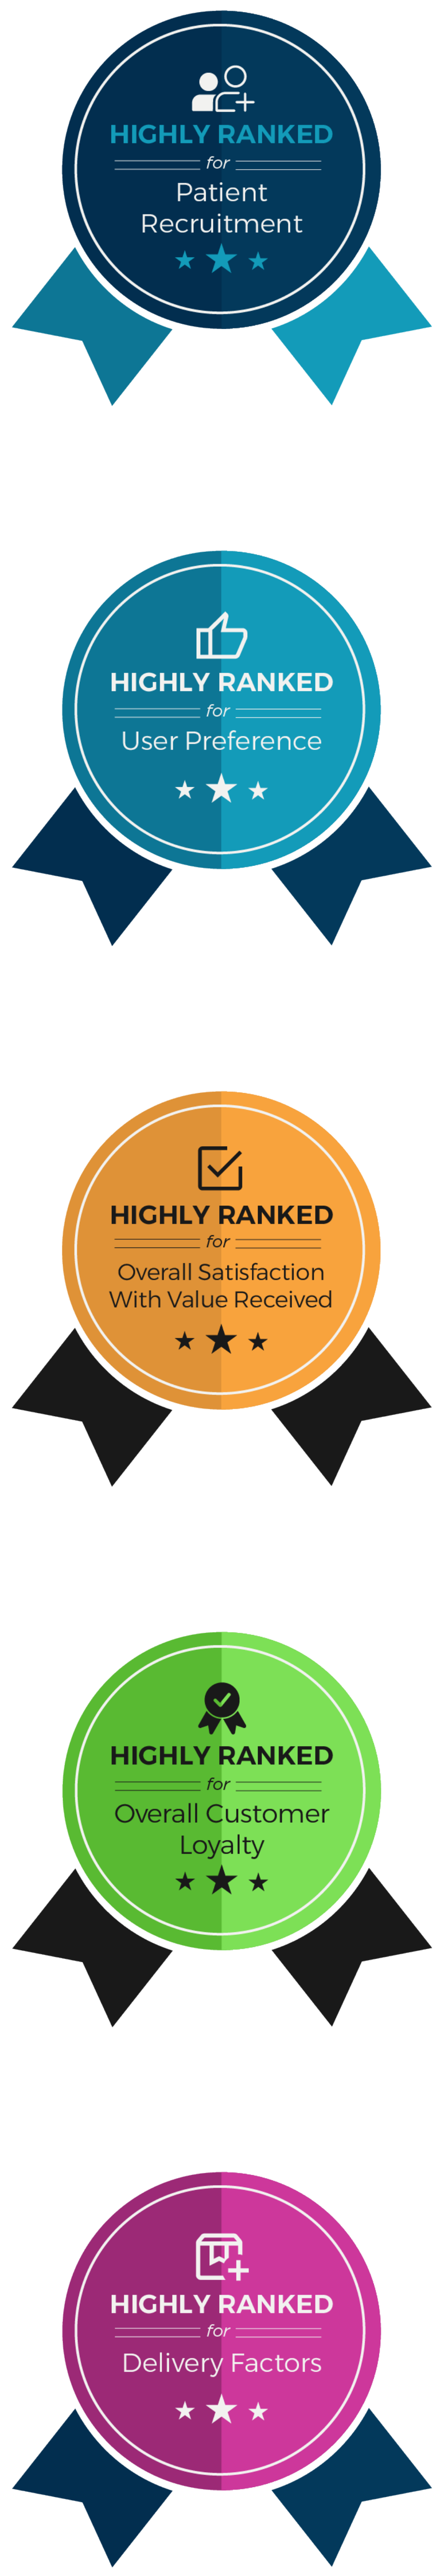

HIGHLY RANKED
for
Patient Recruitment
HIGHLY RANKED
for
User Preference
HIGHLY RANKED
for
Overall Satisfaction With Value Received
HIGHLY RANKED
for
Overall Customer Loyalty
HIGHLY RANKED
for
Delivery Factors

Updated Guidance for Industry
Obesity and Overweight, FDA, Jan 2025

- Reframing obesity as a "chronic disease" (vs. chronic health risk)
- To be "effective", decrease total body weight ≥ 5% in 1 y
- Secondary efficacy: BP, lipids, glucose, and A1c (diabetes)
- Waist circumference is not a surrogate for visceral fat
- Phase 1 sho uld enroll "broad range" of BMI
- Phase 2 should show (near) maximal weight loss
- Phase 3 (1+ study) must include SoC diet/exercise program
- Safety database of 3,000+ (DRUG) vs. 1,500+ (PBO) for 1 y
- Fixed combination drug product (FCDP) must compare efficacy with individual components for (near) maximal weight loss
- Clinical Outcome Assessments / planned analyses should be discussed (i.e., EOP2, Type C)
- For functional assessments, population must have sufficient limitation in functioning to observe a clinically meaningful within-patient change

Choosing the Right Assessment
Meaningfulness vs. Complexity

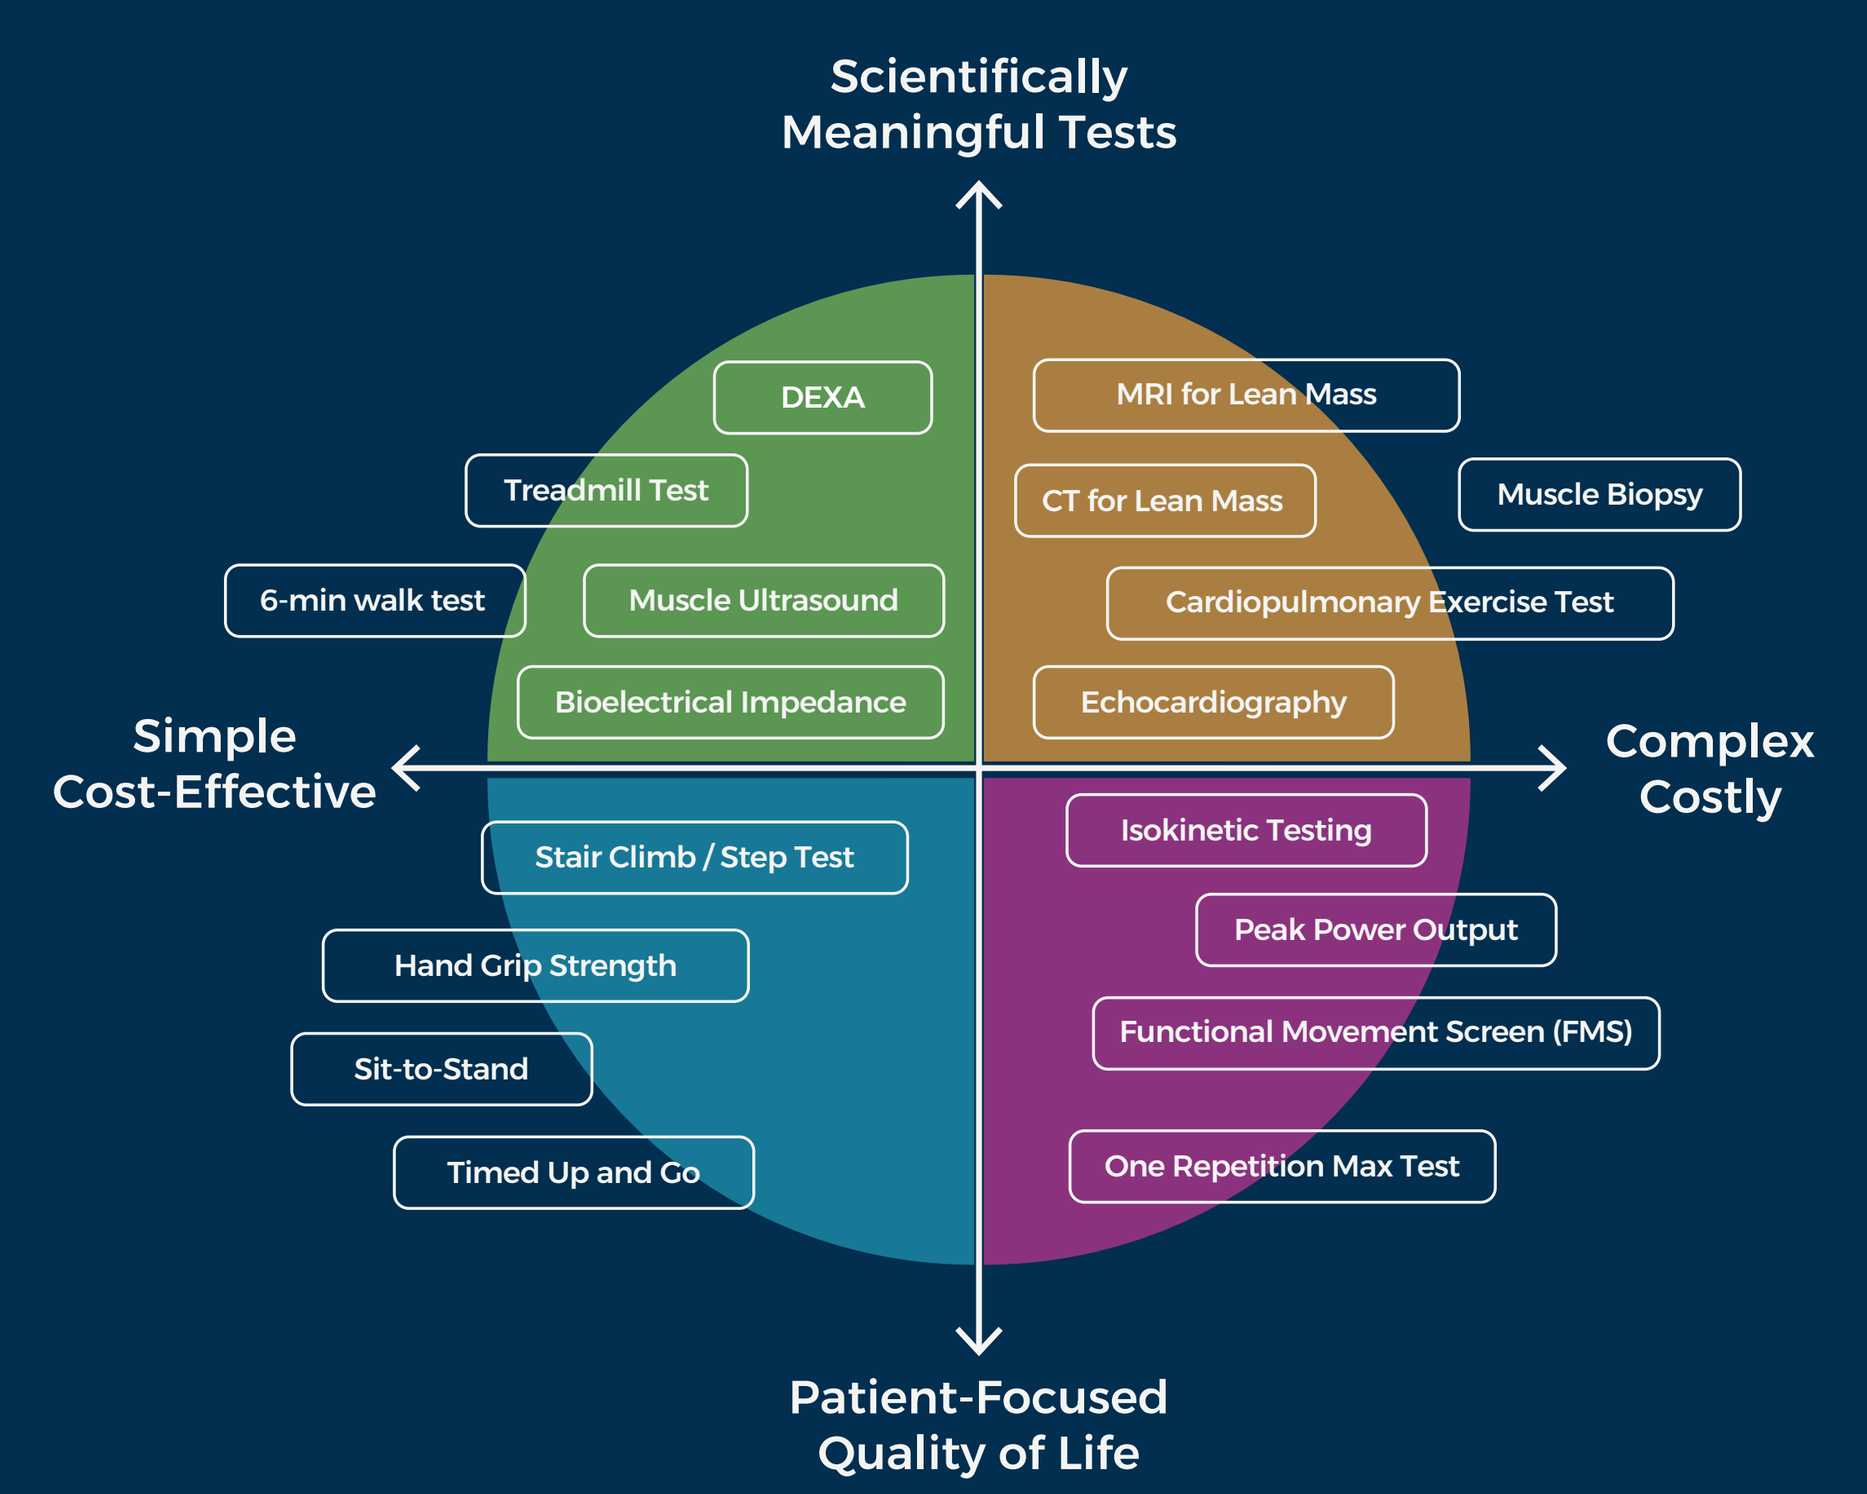

Scientifically Meaningful Tests
Simple Cost-Effective
Complex Costly
Patient-Focused Quality of Life
DEXA
Treadmill Test
6-min walk test
Muscle Ultrasound
Bioelectrical Impedance
MRI for Lean Mass
CT for Lean Mass
Muscle Biopsy
Cardiopulmonary Exercise Test
Echocardiography
Stair Climb / Step Test
Hand Grip Strength
Sit-to-Stand
Timed Up and Go
Isokinetic Testing
Peak Power Output
Functional Movement Screen (FMS)
One Repetition Max Test

Strategic Consulting
Supporting Preclinical to Commercialization

- Portfolio Review: one asset, review of multiple indication(s)
- Clinical Development Plan: P1-3 study designs + ballpark costs
- Synopsis / Protocol Development: ICH-GCP compatible
- KOL Engagement: structured Q&A for design feedback
- Type B (Pre-IND, EOP) Meeting: package and attendance
- Type C / D Meeting: package, attendance, and revisions
- Special Initiatives: FastTrack, Breakthrough, ODD/RPDD
- General Consulting: address specific program questions
- FDA (18 CDER Divisions + CBER), EMA, MHRA, and Health Canada direct liaison experience in the past 5 years

Beyond Weight and BMI
Body Composition and Functional Assessments

Methods
- TrialTrove survey for MeSH term "obesity"
- 5,281 trials in obesity catalogued
- 1,728 trials with 1+ assessment (BMI) used in analysis
- Lookback: 1998-Present
- Status: Past (1,117), Present (334), Future (277)
- Phase: I (87), II (327), III (301), IV (765)

Summary
- Since 2015, the use of 1+ body composition assessment (i.e., DEXA) has remained consistent (~60%).
- However, the proportion of studies with 1+ functional assessments (i.e., walk test) has doubled since 2020.
- This highlights the burgeoning demand for functional assessments within a modern clinical development plans for obesity.

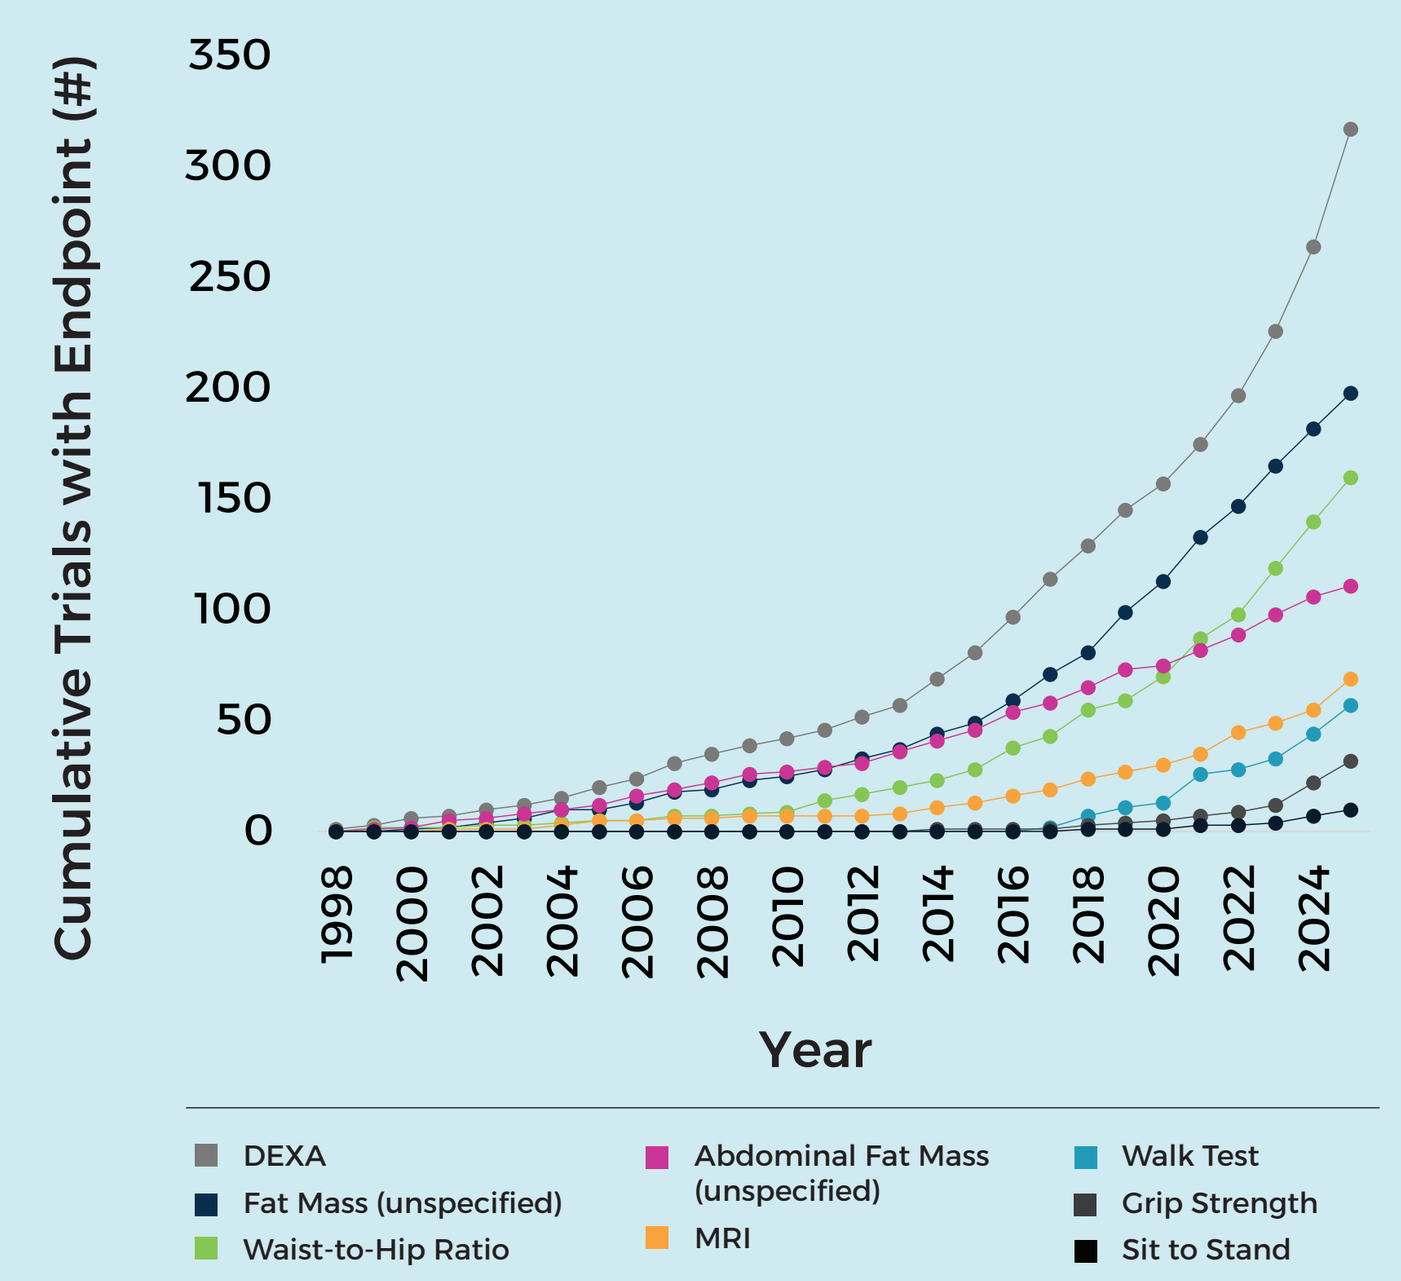

Cumulative Trials with Endpoint (#)
350
300
250
200
150
100
50
0
1998
2000
2002
2004
2006
2008
2010
2012
2014
2016
2018
2020
2022
2024
Year
DEXA
Fat Mass (unspecified)
Waist-to-Hip Ratio
Abdominal Fat Mass (unspecified)
MRI
Walk Test
Grip Strength
Sit to Stand

Evolution of Body Composition Assessments
Measuring Fat Loss Specifically

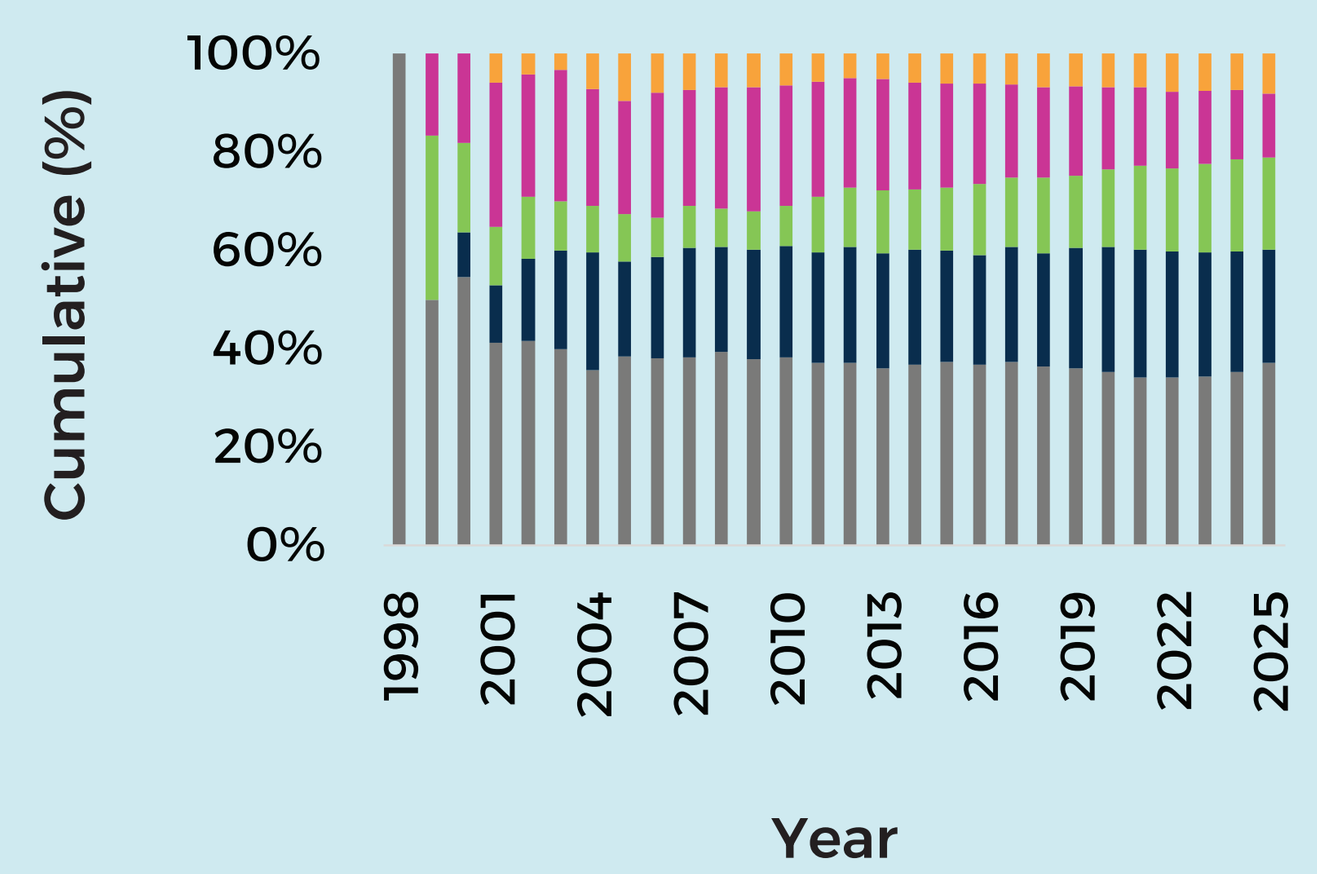

Cumulative (%)
100%
80%
60%
40%
20%
0%
1998
2001
2004
2007
2010
2013
2016
2019
2022
2025
Year

- ASSESSMENTS: DEXA, Fat Mass (unspecified), Waist-to-Hip Ratio, Abdominal Fat Mass (unspecified), MRI
- Most obesity trials (~60%) assess body composition in addition to total weight loss / BMI alone.
- DXA remains the most common assessment of body composition (≥ 40%) compared to MRI (≥ 8%), ultrasound, and bioelectrical impedance.
- Waist circumference and waist-to-hip ratio (≥ 20%) can provide increased sensitivity within shorter duration treatment periods.
- Although MRI (≥ 8%) is more precise, operational complexity and cost limits its utilization in research compared to other assessments.

Emergence of Functional Assessments
Weight Loss with Purpose

- ASSESSMENTS: Walk Test, Exercise Capacity (unspecified), Grip Strength, Sit to Stand, Stair Climb
- Only 2 studies (2004, 2009) were identified with functional assessments prior to 2014 measuring Exercise Capacity (unspecified).
- Grip strength is the most common functional assessment in recent years (≥ 50%), requiring few but specific operational requirements.
- Variability in Walk Test methods make interstudy comparison challenging (i.e., 6-Minute, Timed Up and Go, Shuttle Walk).
- Sit to Stand and Stair Climb tests are gaining popularity, aiming to provide a holistic assessment of patient function and quality of life.

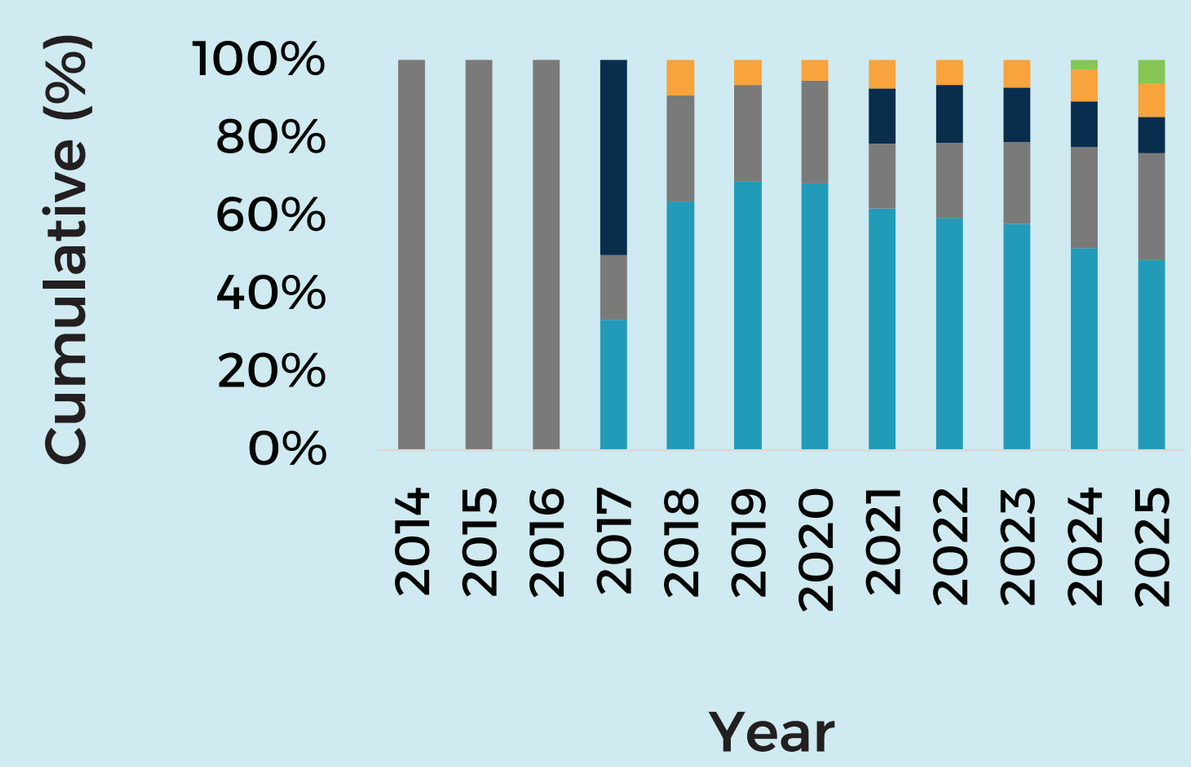

Cumulative (%)
100%
80%
60%
40%
20%
0%
2014
2015
2016
2017
2018
2019
2020
2021
2022
2023
2024
2025
Year

Making It Count
Operational Considerations to Maximize Precision

Body Composition and Distribution	
DXA	• Experienced technician / central reading • Patient preparation: positioning and immobilization • Weight limits per machine, need for alternative sites
MRI	• Experienced technician / central reading • Equipment calibration (i.e., phantoms) • Variable Specific Absorption Rate (SRA) • Timely data assessment and upload • Safety screening (i.e., metallic implants) • Patient preparation: closed space, loud noise, duration • Adjustments for body size, position, and comfort
CT	• Experienced technician/central reading • Radiation Safety Approval (ionizing radiation exposure) • Adjustment for clothing and artifact • Contrast / pre-medication • Patient preparation: positioning and immobilization
Bioimpedance	• Derived measures: age, gender, ethnicity, and activity • Fasted state and exercise limitation • Varies by fluid balance (no alcohol/caffeine) • Screening (i.e., pacemaker, cochlear implant) • Patient preparation: positioning • Diurnal acquisition timing for max reproducibility

Body Composition and Distribution	
Hand Grip Strength	• Calibration of equipment • Handle size selection for max reproducibility • Proper shoulder, elbow, and forearm positioning • Patient preparation: hand dominance, removal of jewelry, and medical condition considerations (i.e., prior injury, carpal tunnel, arthritis) • Diurnal acquisition timing for max reproducibility • Repeat measures in order of testing, avoid fatigue
Stair Climb Power Test	• Controlled environment with standardized course • Baseline measurement • Collection of vital signs throughout • Staff training with standardized verbal cues • Diurnal acquisition timing for max reproducibility • Patient preparation: clothing, shoes, mobility aid • Tester training: scoring criteria, safety, timing, data collection/recording, qualitative observations • Emergency Stop Procedures
Sit to Stand	• Controlled environment with standardized course • Patient preparation: practice trials, starting position • Patient preparation: clothing, shoes, mobility aid • Tester training: positioning, safety, timing, data collection/recording, qualitative observations • Emergency Stop Procedures
Walk Test	• Controlled environment with standardized course • Baseline measurement • Collection of vital signs throughout • Staff training with standardized verbal cues • Validated scale, stopwatch, BP, Pulse Ox, AED • Diurnal acquisition timing for max reproducibility • Patient preparation: clothing, shoes, mobility aid • Emergency Stop Procedures

Contact Us

- Complimentary Study Design Q&A / Synopsis Review
- Request for Information (RFI)
- Request for Proposal (RFP)

Ask us about our experience in Obesity, T2D, and MASH.

- Peyton.Sandroni@Worldwide.com
- MarieElena.Cordisco@Worldwide.com
- Natalia.Castro@Worldwide.com