A Plate-based, Automated Liquid/Liquid Extraction (LLE) Method for Improving Operational Efficiency of LC-MS/MS Methods

Leimin Fan, Alicia Bian, and JP Kotha
Southpark Commerce Conter Two, 4500 Fredrich Lane, Suit 200, Austin, TX 78744, USA
Worldwide Clinical Trials

Worldwide Clinical Trials

18th WRIB
Where Regulators & Industry Convene

Novel Aspects:

Modification of a tube-based liquid/liquid extraction (LLE) method to a **48-well plate** - automated liquid handler method for analyzing compound A in human K_2-EDTA plasma for improving efficiency and data quality. This modification allows to overcome volume limitation of using a 96-well plates in LLE methods.

Introduction:

Liquid-liquid extraction (LLE) 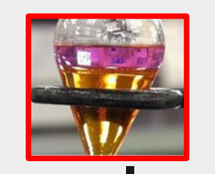 is a highly effective technique for extracting compounds in bioanalysis. The LLE method involves transferring an analyte of interest between an immiscible organic solvent and an aqueous phase. To selectively extract desired analytes with sufficient recovery, a large volume of immiscible organic solvents like hexane, MTBE, or Ethyl Acetate are commonly used in LLE methods. Oftentimes, the total extraction volume of an LLE method exceeds the capacity of a single well in a 96-well plate 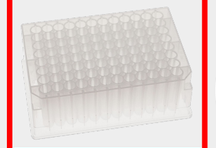 (typically 1.5mL usable volume in a 2-mL plate). Conventionally, 12x75 mm glass culture tubes 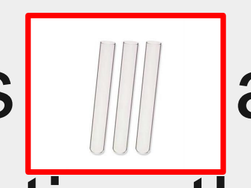 are used to accommodate larger volume of solvents during extraction that require manual processes for organic phase transfer, dry-down, and reconstitution. Unfortunately, such manual methods during LLE involves handling many tubes significantly increasing overall time for the extractions and the risk of exposure to hazardous material. Furthermore, this laborious process can cause repetitive strain injuries to technologists performing these methods on regular basis.

Automated LLE methods for measuring drug levels in biologic fluids using 96-well plate have been reported since the introduction of liquid handlers in bioanalysis workflow. However, when analytical methods require a larger extraction volume, manual LLE methods using tubes are often employed to overcome volume limitations posed by 96-well format . In this research, we employed a modified LLE method by using 48-well plates 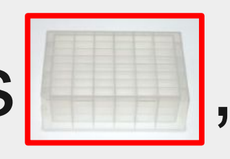, Hamilton Star 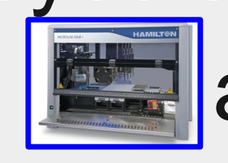 and Tomtec Quadra 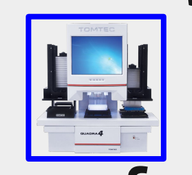 liquid handlers. A single well in a 48-well plate has a useful volume of more than 4 mL that is sufficient for most LLE methods. In this modified method, Hamilton system was used for aliquoting and preparation of internal standards and a Tomtec system was used for transferring extraction reagents and the organic phase. Hamilton Star was also employed for sample transfer after reconstitution. This modified LLE method notably improved the efficiency and performance of the assay.

Method:

The analytical method applies a standard liquid/liquid extraction using Hexanes/Isopropanol (9:1) as the organic extraction solvent. The assay range is 0.1 to 200 ng/mL of Compound A in human K_2-EDTA plasma. The QC samples were prepared at Compound A concentrations of 0.3, 10, 100, and 160 ng/mL. The steps for LLE method are outlined below.

Extraction:

1. Add Samples (0.2 mL) into 48-well polypropylene plates (Stellar Scientific 48-well U bottom Extraction Plate) using a Hamilton Star liquid handler.
2. Add deuterium labeled internal standard working solution (20 µL).
3. After a brief mixing, add 2.5 mL of Extraction Solution to the wells using Tomtec Quadra 4 liquid handler.
4. Heat seal the plates, vortex, and centrifuge for phase separation.
5. Transfer top organic layer into Corning® Axygen® 48-Well Pyramid bottom plates and evaporate the organic sample to dryness under a gentle stream of nitrogen at ~40°C.
6. Reconstitute with 0.15 mL of acetonitrile/water/formic acid solution (60:40:0.025) and and vortex at 1500 rpm on MixMate for approximately two minutes.
7. Transfer reconstituted samples into glass insert vials in a 96-position carrier plate and seal with a capmat.
8. Analyze the samples by a **Sciex API 5000** LC-MS-MS system equipped with an HPLC column.

LC-MS/MS analysis:

Column: Kinetex 2.6 µm XB-C18, 2.1 x 50 mm, Phenomenex, 00B-4496-AN
Mobile Phase A: 1 mM ammonium trifluoroacetate in water
Mobile Phase B: Acetonitrile/Isopropanol; 9:1
Flow Rate: 0.4 mL/min
Injection Volume: 10 µL

Typical Chromatograms:

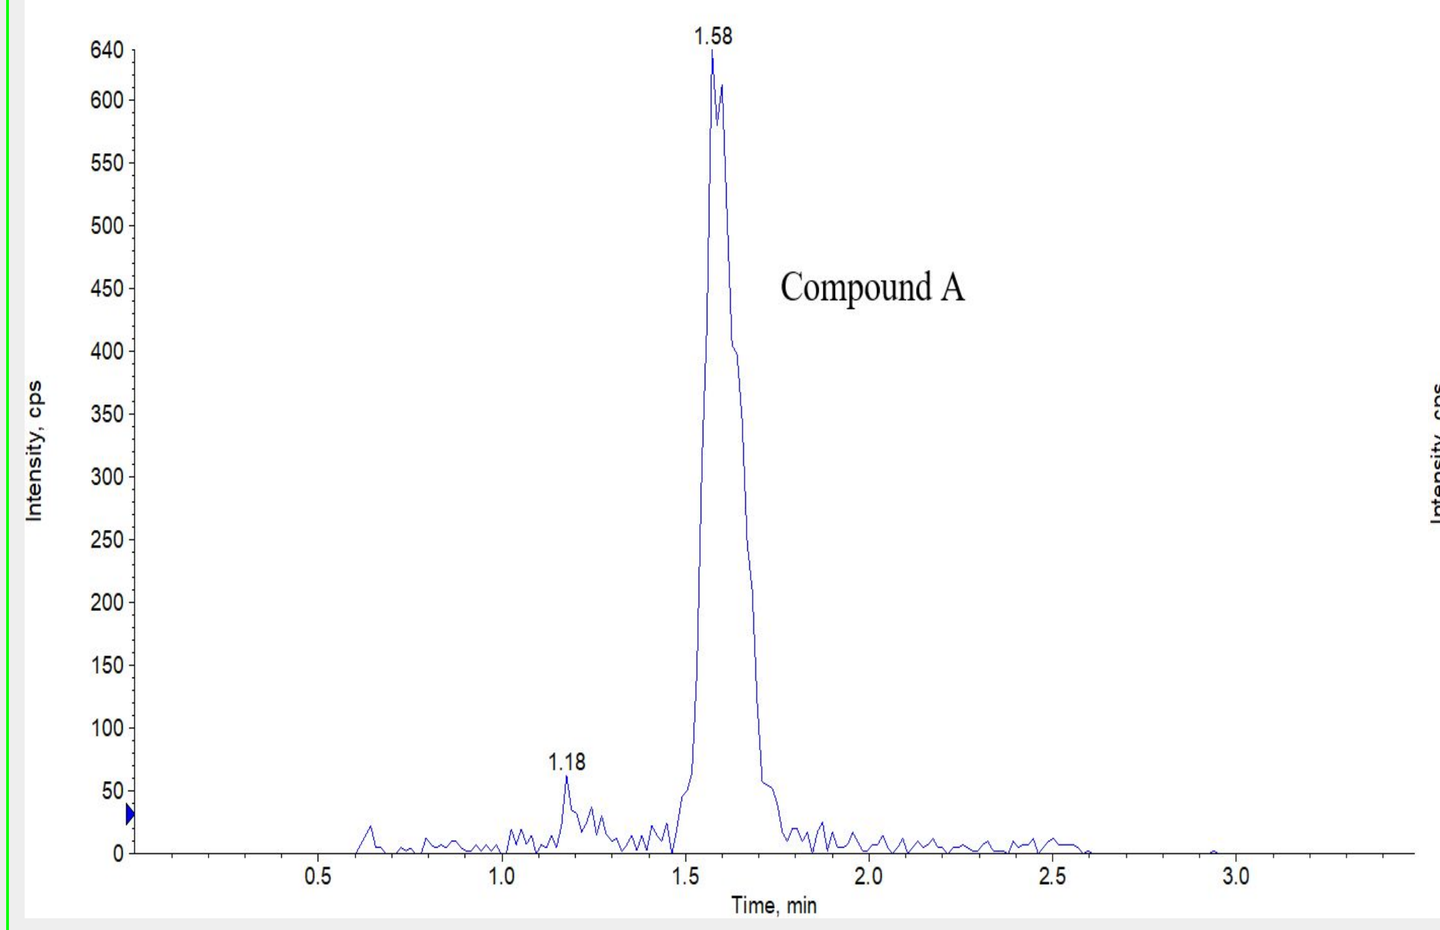
1.58
Compound A
1.18
Intensity, cps
Time, min
LLOQ

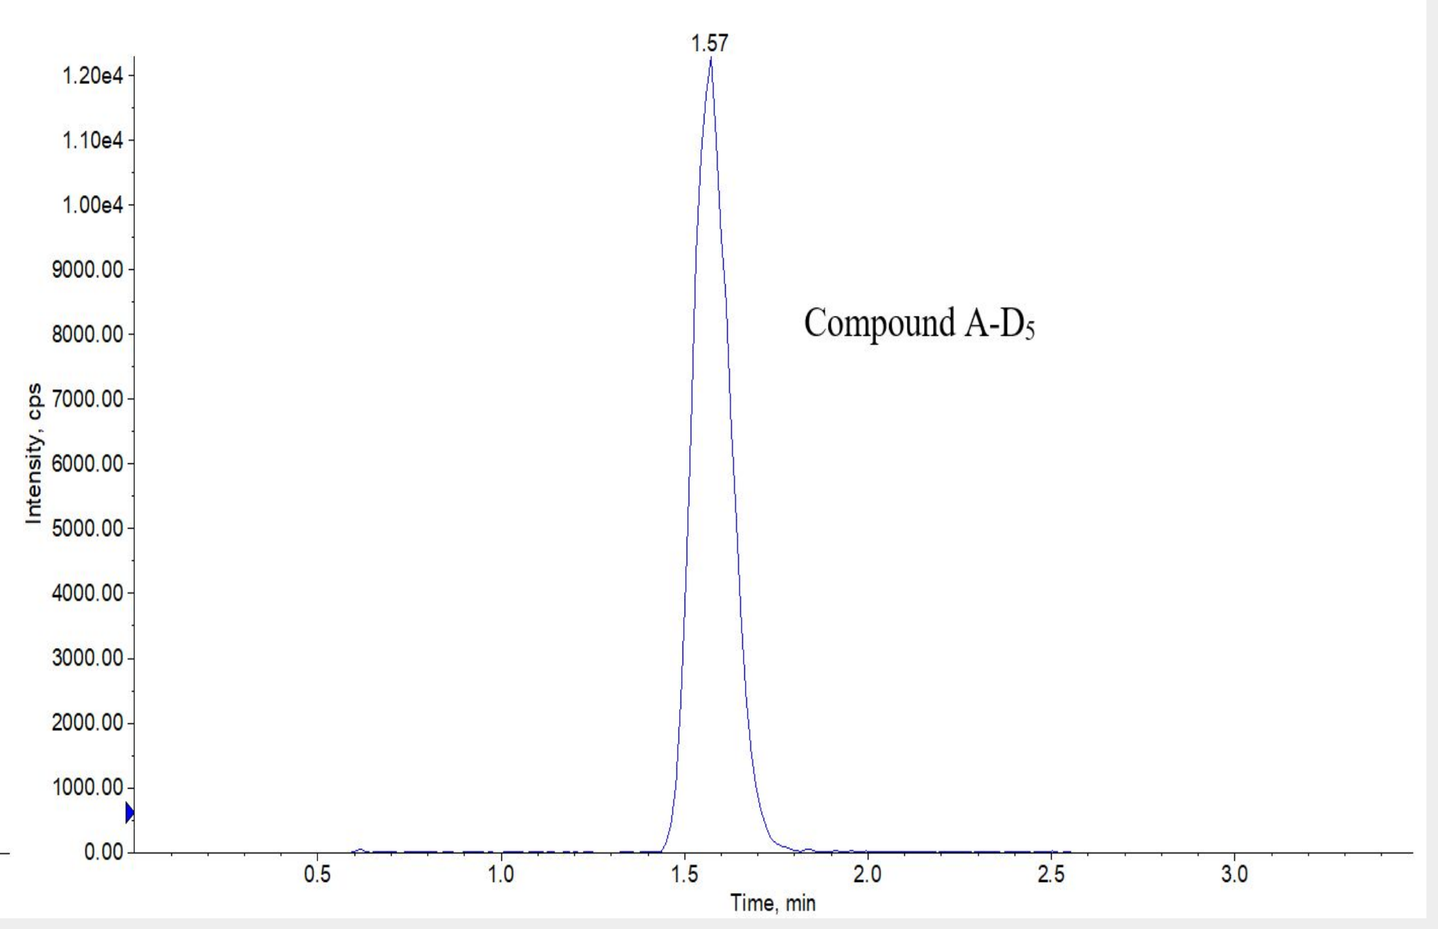
1.57
Compound A-D5
Intensity, cps
Time, min
Internal standard

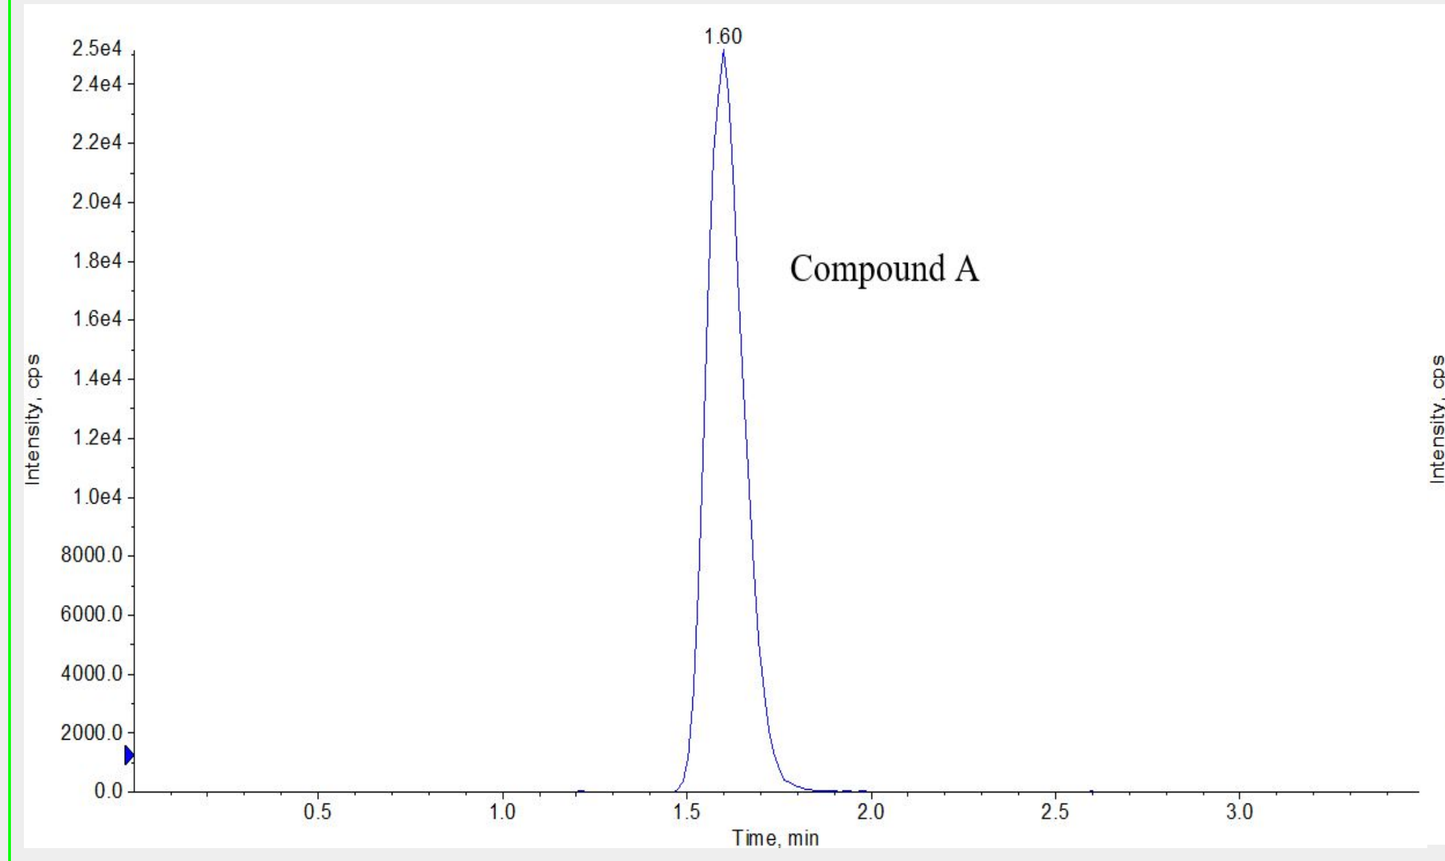
1.60
Compound A
Intensity, cps
Time, min
Standard D, 5.0 ng/mL

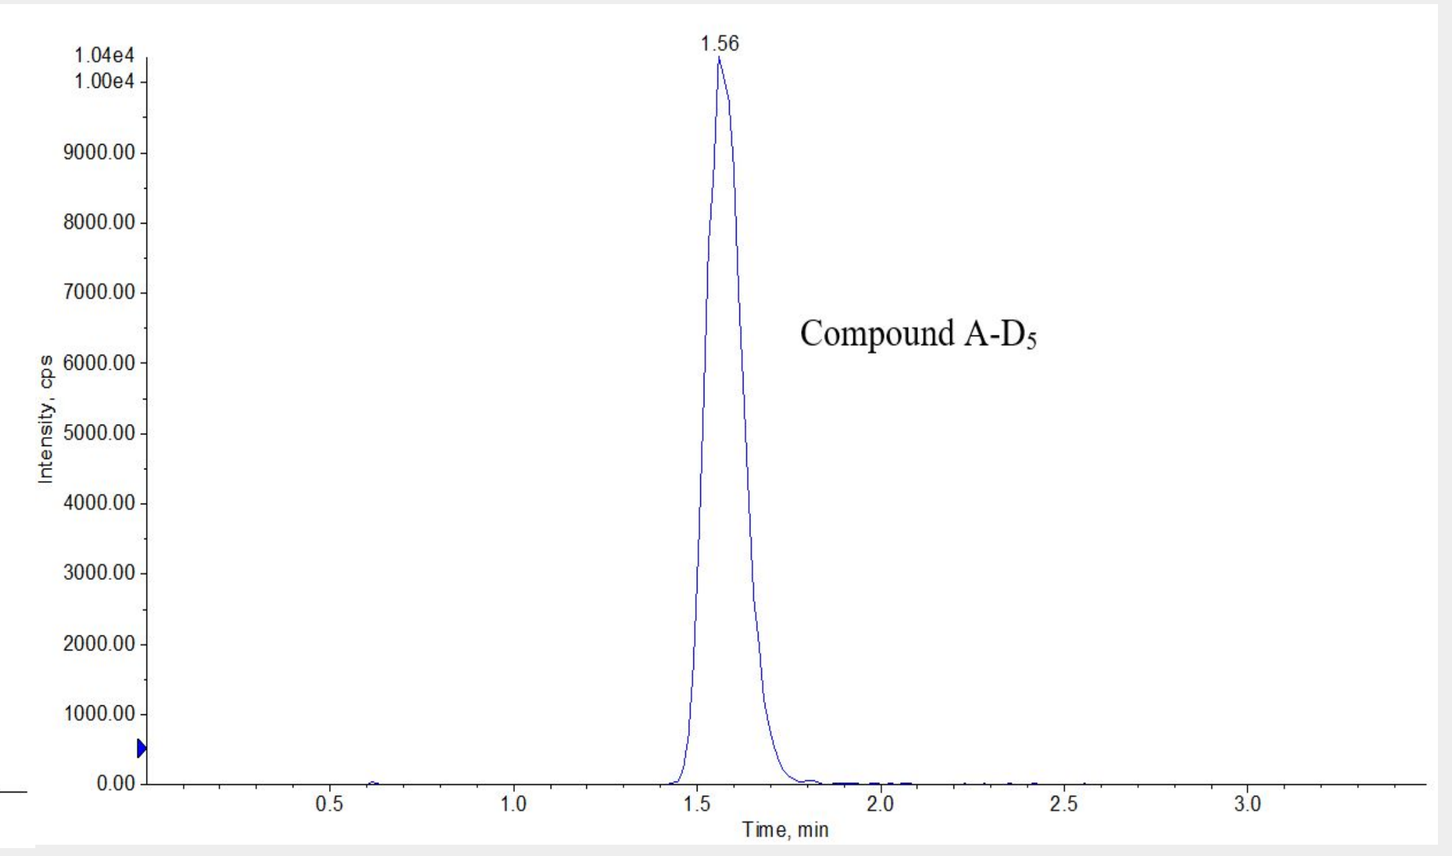
1.56
Compound A-D5
Intensity, cps
Time, min
Internal standard

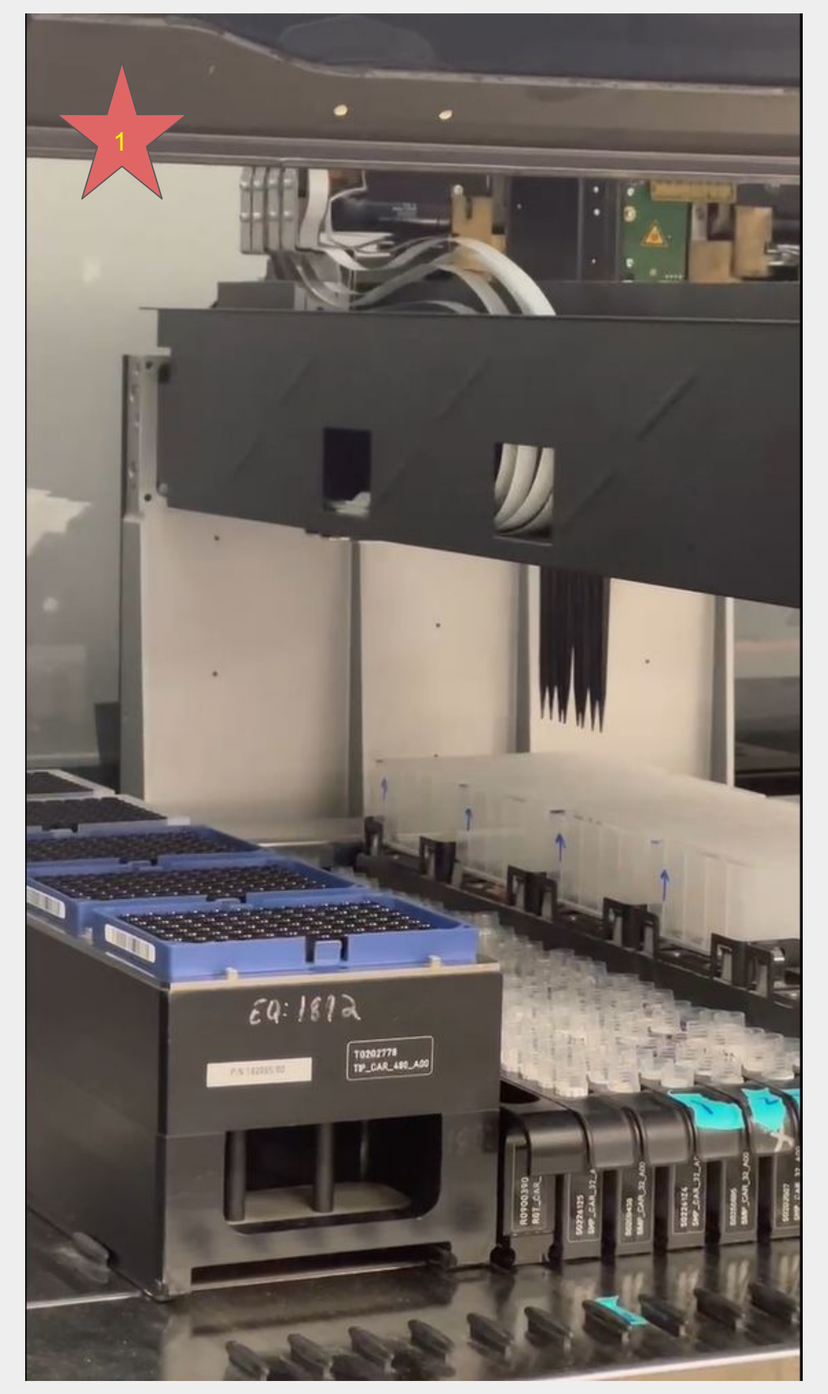
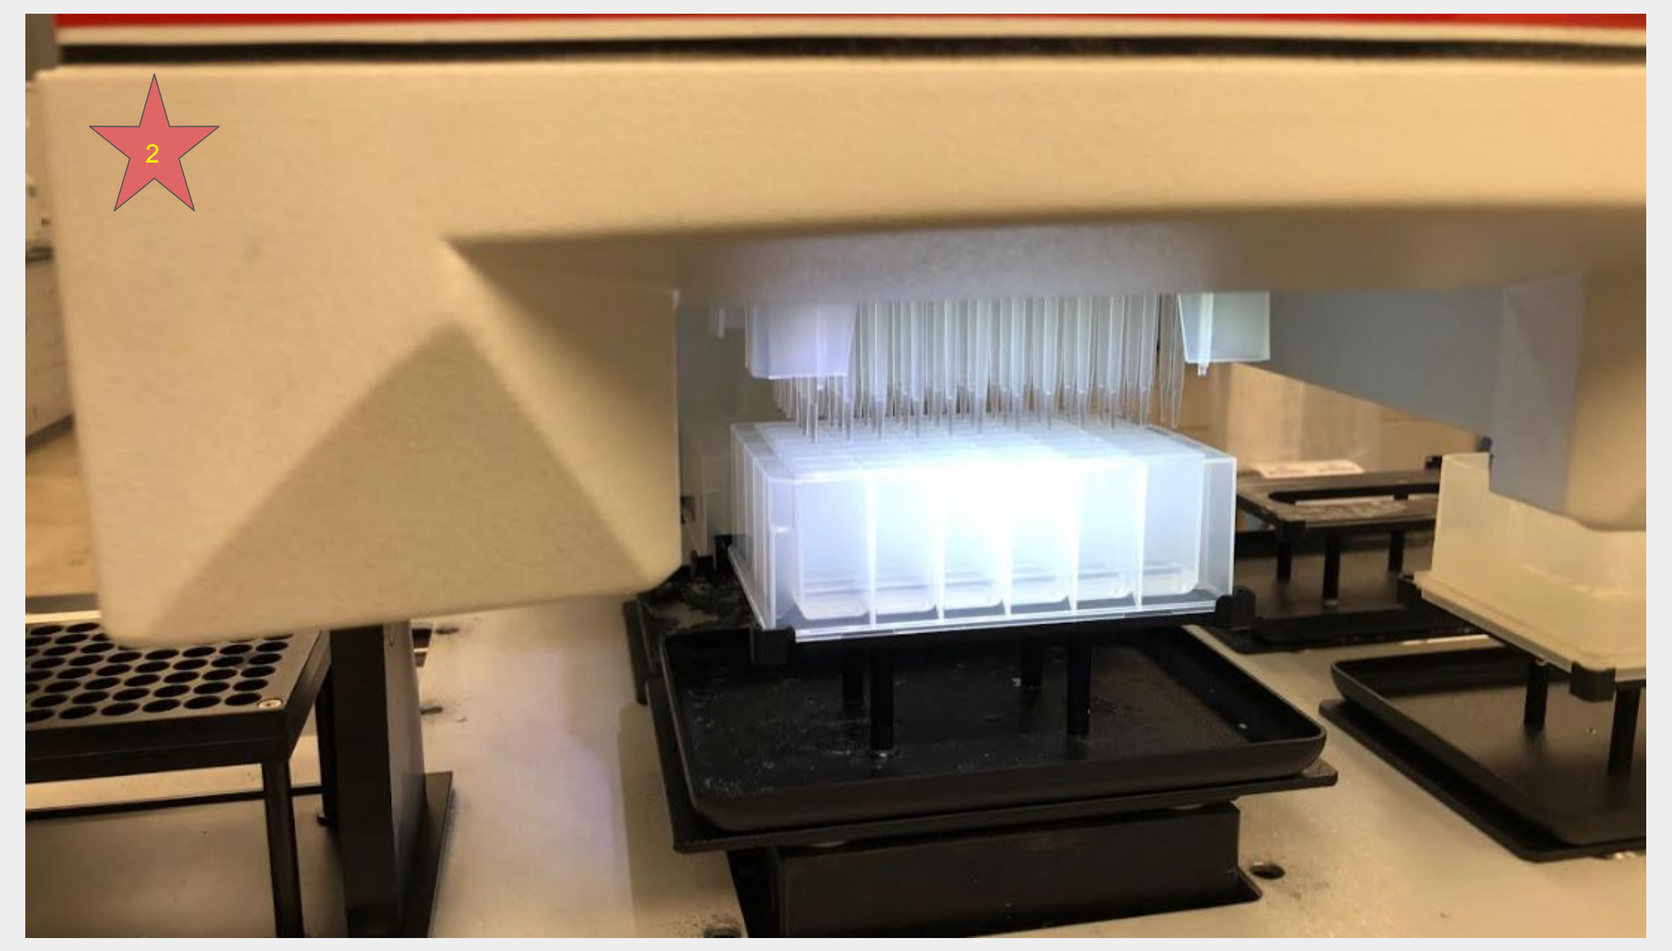
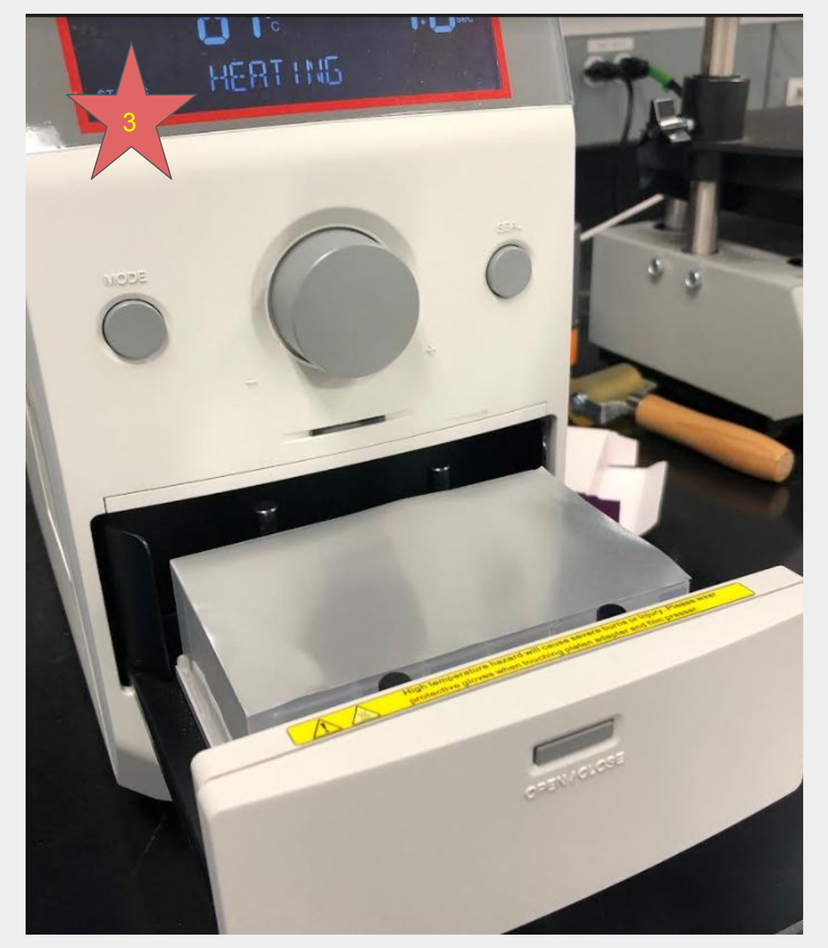
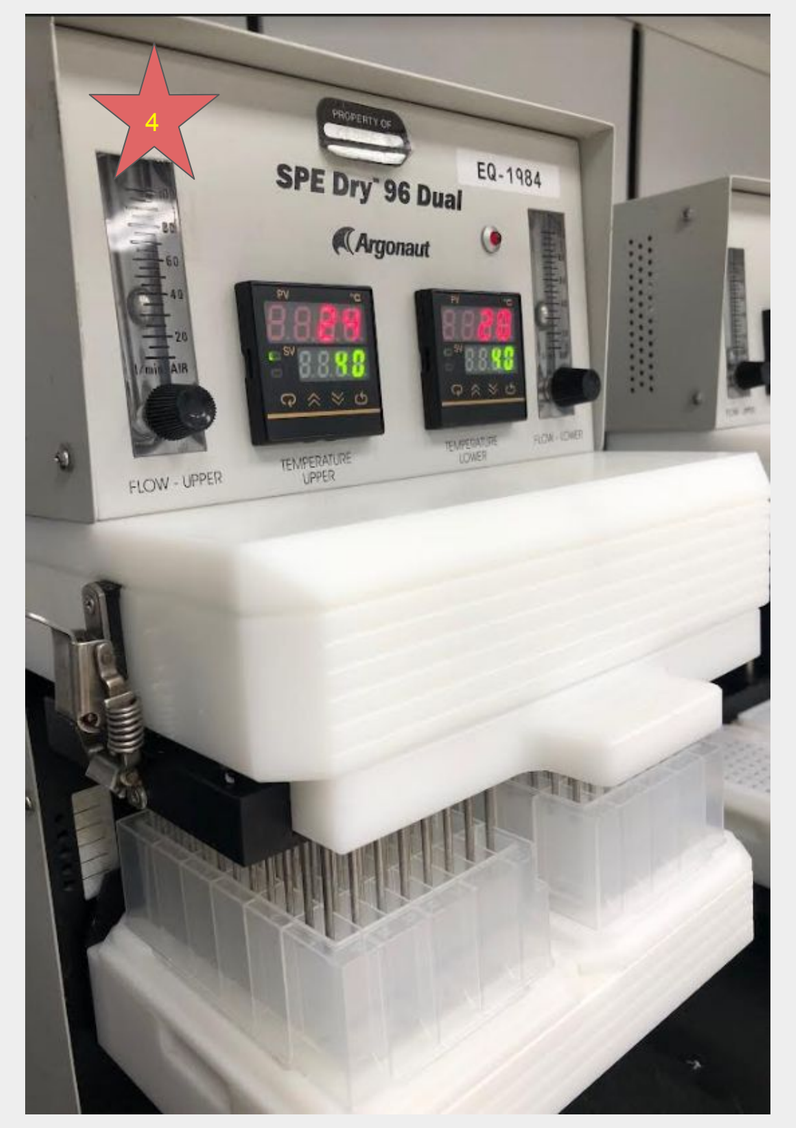

1. Aliquot samples with Hamilton StarLet
2. Organic Transfer by Tomtec
3. Heatseal
4. Dry-down

Results:

- The method demonstrated exceptional ruggedness. It was successfully applied to a clinical trial with over 5200 samples.
- Due to simplified process without significant manual operation, fast turnaround time of 3 days data delivery over 1000 samples each time was achieved.

Standard Curve Performance

	A	B	C	D	E	F	G	H	I	J	K	L
2	Concentration [ng/mL]	A 0.100 ng/mL	B 0.200 ng/mL	C 1.00 ng/mL	D 5.00 ng/mL	E 20.0 ng/mL	F 100 ng/mL	G 180 ng/mL	H 200 ng/mL	Slope	Intercept	R-Squared
3	Batch 2	0.100	0.199	0.984	5.34	20.9	99.5	173	189	0.846662068	0.009026128	0.9980
4	Batch 3	0.102	0.194	0.986	4.87	20.8	98.3	187	199	0.924210331	0.007685406	0.9991
5	Batch 1	0.100	0.199	0.988	5.30	20.4	101	174	191	0.929905768	-0.005820131	0.9987
6	Batch 5	0.0970	0.213	0.974	5.15	20.6	101	175	189	0.799405192	0.010217244	0.9981
7	Batch 6	0.101	0.199	0.945	5.15	21.4	96.1	182	196	0.600230987	-0.001623565	0.9982
8	Batch 7	0.101	0.198	1.01	5.05	20.3	101	175	198	0.477820512	0.002713659	0.9998
9	Batch 10	0.0998	0.201	0.991	4.99	20.3	98.9	181	200	0.586271138	-0.006566188	0.9999
10	Batch 8	0.102	0.192	0.987	5.00	20.2	101	178	205	0.810926622	-0.001002368	0.9994
11	Batch 9	0.0995	0.202	0.995	5.20	20.6	99.9	177	190	0.873113951	0.005878199	0.9991
12	Batch 11	0.0992	0.203	1.00	5.22	20.2	98.7	175	196	0.461168262	0.001556025	0.9994
13	Batch 12	0.0978	0.207	1.04	5.03	20.6	104	179	176	0.473935301	-0.000121776	0.9966
14	Batch 13	0.102	0.192	1.01	5.12	20.6	102	174	195	0.457409982	0.008288839	0.9991
15	Batch 14	0.0986	0.207	0.968	4.91	20.4	98.1	181	205	0.806105890	-0.002482830	0.9993
16	Batch 15	0.102	0.193	0.988	5.19	20.6	99.7	179	194	0.671867767	0.005521596	0.9992
17	Batch 16	0.0996	0.202	0.982	5.12	19.7	104	175	198	0.492581768	0.004595398	0.9994
18	Batch 17	0.0972	0.211	*No Value	5.36	20.6	97.4	166	196	0.727142994	0.001093950	0.9967
19	Batch 18	0.0990	0.202	1.04	5.10	21.1	98.1	168	193	0.428532459	0.003437928	0.9981
20	Batch 20	0.0986	0.206	0.987	5.00	20.3	99.5	181	196	0.592499378	0.003193874	0.9997
21	Batch 19	0.101	0.196	1.00	5.11	20.6	103	173	193	0.580884332	0.002493056	0.9991
22	Batch 21	0.0983	0.206	1.01	4.98	19.8	99.0	183	197	0.514728117	0.003465741	0.9996
23	Batch 22	0.0959	0.217	0.995	4.98	19.7	101	180	194	0.504728637	0.004466133	0.9983
24	Batch 23	0.0996	0.203	0.972	5.07	20.1	101	178	201	0.550489819	-0.004071856	0.9998
25	Batch 25	0.0976	0.205	1.09	5.53	21.6	97.3	158	175	0.830324419	0.009793169	0.9904
26	Batch 24	0.0998	0.200	1.00	5.03	20.1	100	179	197	0.553341249	-0.003006276	0.9999
27	Batch 26	0.0921	0.228	1.08	5.40	19.5	98.8	158	188	0.835279200	0.002903031	0.9901
28	Batch 27	0.0991	0.202	1.02	5.16	20.8	101	168	191	0.518365733	0.006249615	0.9984
29	Batch 28	0.0995	0.198	1.09	5.62	21.8	90.7	164	180	0.840550134	0.011826310	0.9902
30	Batch 30	0.100	0.198	0.999	5.17	20.7	101	173	194	0.509213149	-0.001309268	0.9992
31	Batch 31	0.101	0.194	0.996	5.11	19.5	102	175	205	0.640437335	-0.004515201	0.9993
32	Batch 35	0.0986	0.205	1.02	5.15	20.9	98.8	168	195	0.696279351	-0.003047327	0.9985
33	Batch 32	0.0998	0.196	1.10	5.45	21.8	99.8	161	171	0.594966694	0.002760617	0.9903
34	Batch 33	0.0981	0.208	1.01	4.88	20.6	103	167	202	0.623601505	0.002810213	0.9984
35	Batch 36	0.0987	0.203	1.05	5.02	20.6	97.0	176	193	0.421612144	0.001591606	0.9988
36	Batch 37	0.0981	0.206	1.03	5.26	20.7	100	170	186	0.465342796	-0.001865811	0.9977
37	Batch 38	0.0985	0.205	1.02	5.27	19.9	99.3	174	194	0.594825559	-0.002450120	0.9989
38	Batch 39	0.101	0.197	1.02	4.86	19.7	103	179	201	0.539690138	-0.001793849	0.9995
39	Batch 40	0.0998	0.200	1.01	5.09	20.1	101	175	198	0.797572468	0.002287986	0.9998
40	Batch 41	0.105	0.181	**0.823	4.89	19.7	110	169	210	0.575735966	0.001211400	0.9944
41	Batch 42	0.103	0.189	0.969	4.82	20.4	101	187	203	0.753382608	-0.001212493	0.9985
42	Batch 43	0.0979	0.208	1.01	5.15	19.7	98.6	173	203	0.741395437	-0.009689092	0.9991
43	Batch 46	0.0991	0.204	0.972	5.14	19.6	99.7	171	212	0.535475290	-0.004020403	0.9986
44	Batch 47	0.0984	0.204	1.04	5.37	19.9	101	171	186	0.491941920	0.002603635	0.9975
45	Batch 48	0.103	0.186	1.03	5.09	19.5	103	177	200	0.531229634	-0.004666282	0.9985
46	Batch 50	0.102	0.193	0.999	5.25	20.0	102	177	194	0.597300338	0.000975801	0.9991
47	Batch 51	0.0986	0.205	1.00	5.33	19.4	103	174	190	0.474395447	-0.005857991	0.9983
48												
49	n	45	45	43	45	45	45	45	45	45	45	45
50	Overall Mean	0.0996	0.201	1.01	5.14	20.4	100	174	195	0.628286218	0.001189416	0.9980
51	S.D.	0.00213	0.00802	0.0341	0.179	0.604	2.78	6.57	8.40	0.147723000	0.004878436	0.0026
52	%CV	2.1	4.0	3.4	3.5	3.0	2.8	3.8	4.3	23.5	410.2	0.3
53	%Bias	-0.4	0.5	1.0	2.8	2.0	0.0	-3.3	-2.5			
54												
55	Reasons Deactivated											
56	* Technical Error											
57	** >15.0% Bias											

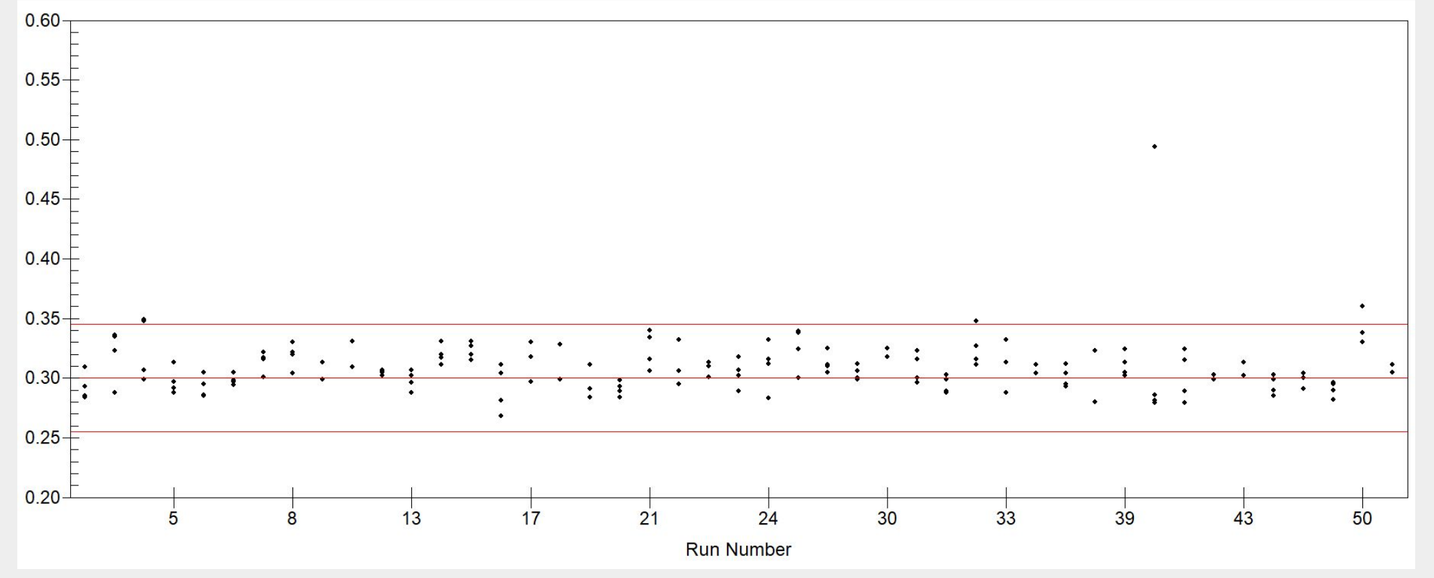
Run Number
QC Low 0.3 ng/mL

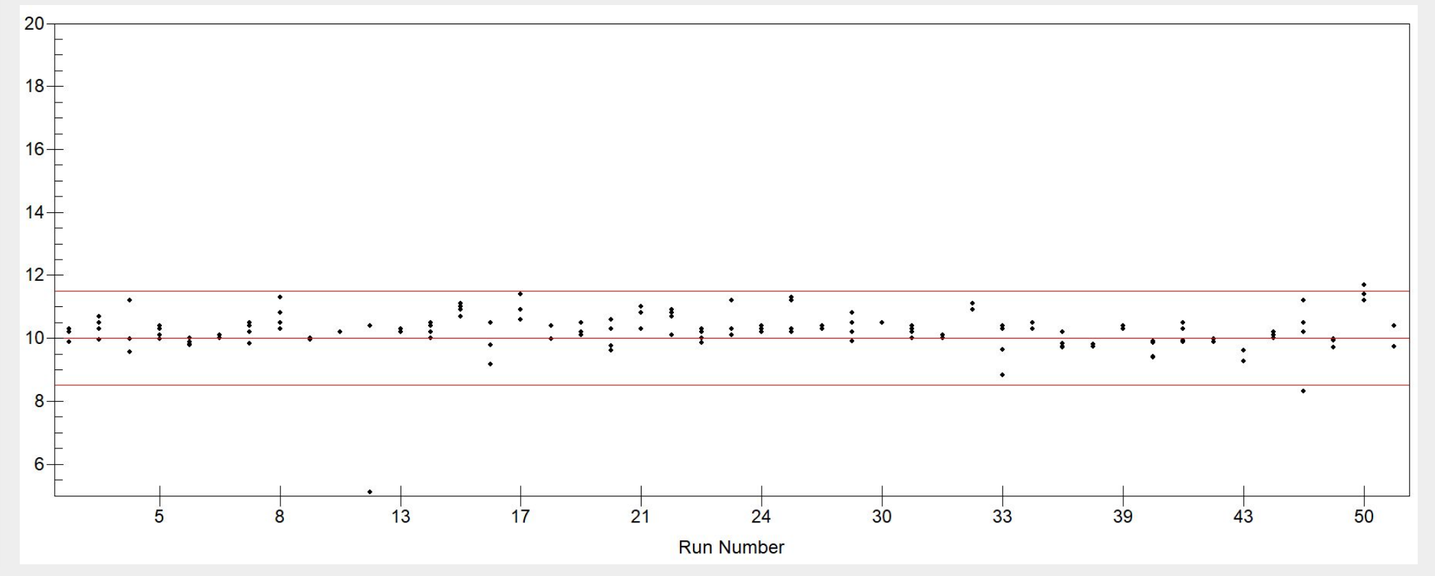
Run Number
QC Mid-Low 7.0 ng/mL

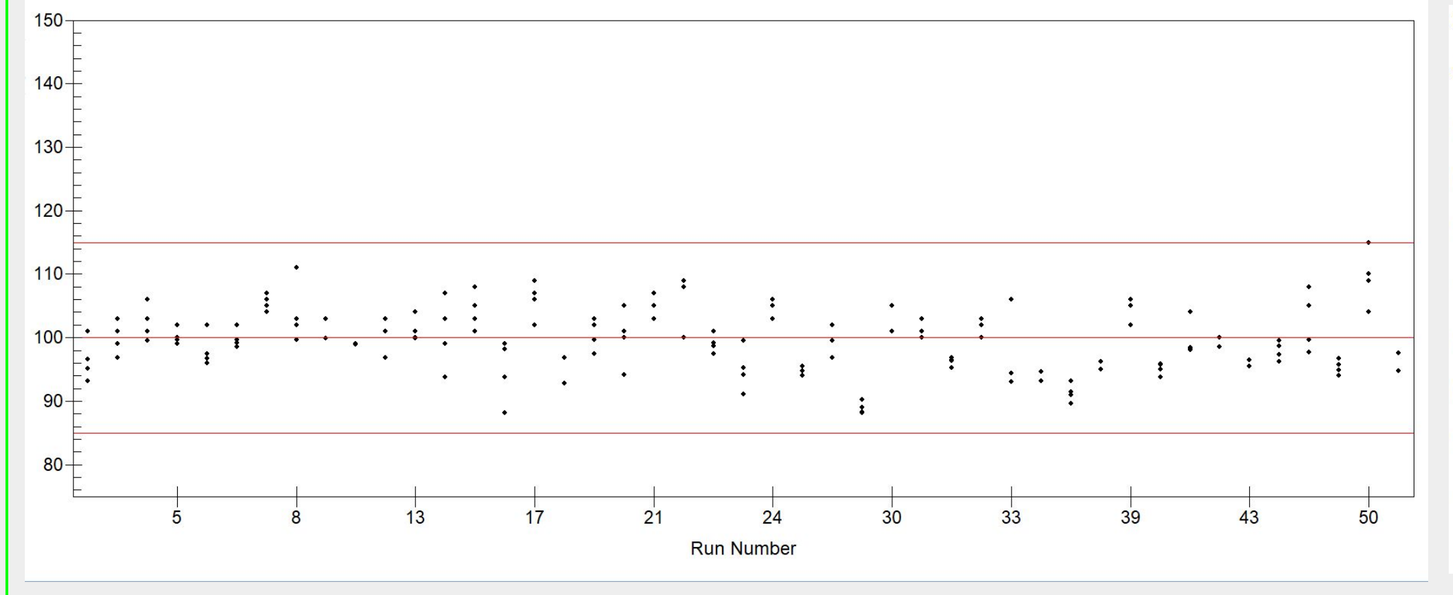
Run Number
QC Mid 100 ng/mL

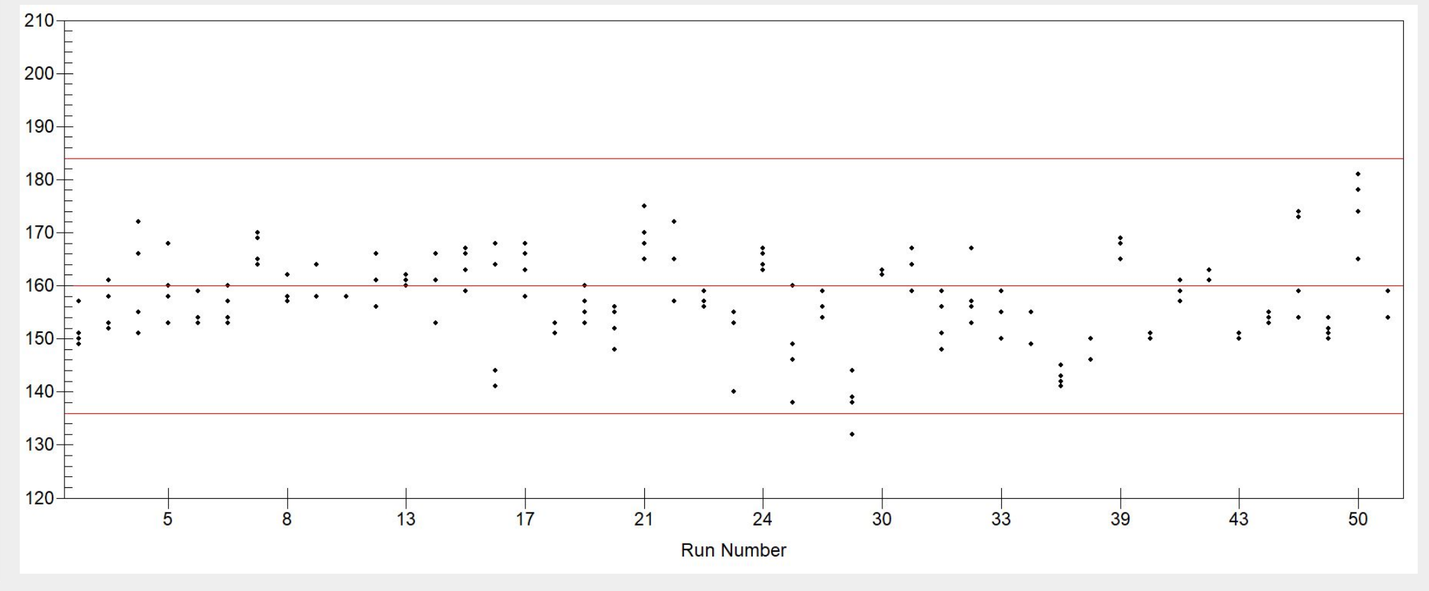
Run Number
QC High 160 ng/mL

ISR Results:

Overall 311 samples were retested for ISR. 13 samples (4.18%) were flagged with % difference >20%.

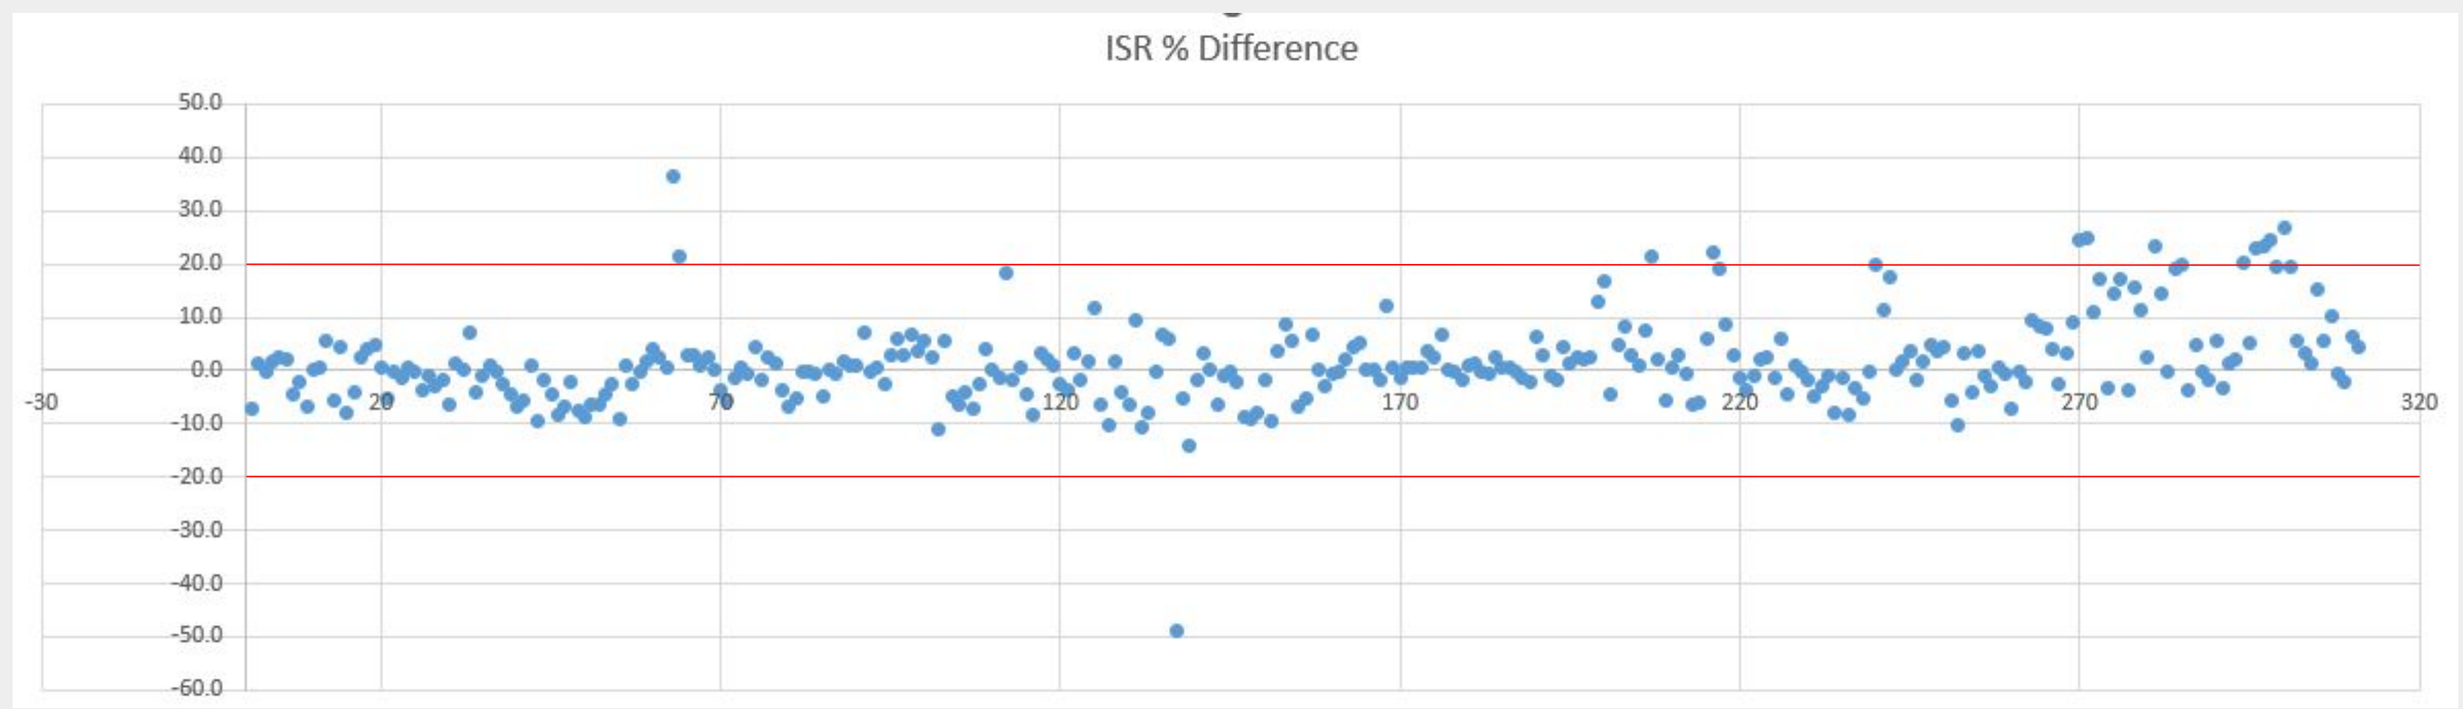
ISR % Difference

Discussion:

Liquid/liquid extraction using automatic liquid handlers for LC-MS/MS methods have been reported for many years. However, for a sample volume> 100 µL, more than 1 mL of organic extraction solution is needed. The volume is far exceeding the total volume of a single well of a 96-well plate. An alternative method of using tubes for extraction using large volumes requires many steps such as tube labeling, capping/decapping, tube arrangement to be performed manually. Here we describe a **48-well** plate format for an LLE method performed using automated liquid handler eliminating manual steps during extraction process. The efficiency to process samples is significantly improved due to this modification. An average skilled scientist can analyze two full batches (192 sample per batch) in an eight-hour shift. With manual steps removed, the assay performance was significantly improved.

When using Tomtec Quadra to transfer liquid, two channels delivery samples to a well so the total transfer volume is programmed in half. Similarly, two channels of the evaporator are applied to a well during the dry-down step.